Neurocognitive Effects of Gluten Exposure: Qualitative Results of a Nationwide Survey

K. Yates [1], J. Edwards George [1], K. Voorhees [2], B. Aideyan [1], K. Sweet [2], J. O'flynn [1], D. Leffler [3], A. Bast [2],*

[1]Northeastern University - Boston, MA (United States), [2]Beyond Celiac - Ambler, PA (United States), [3]Takeda - Cambridge, MA (United States)

*Corresponding author(s) - Email: abast@beyondceliac.org

Background and Objective

Neurocognitive effects after gluten exposure (e.g. "celiac/brain fog") are commonly described by patients with celiac disease (CD) and non-celiac gluten sensitivity (NCGS) though little data exists on the prevalence and specific symptoms associated.

In 2013 a national survey was completed by 1,396 individuals with CD (82%) and NCGS (18%). 89% of CD and 95% of NCGS respondents reported having experienced neurocognitive effects after gluten exposure. These neurocognitive symptoms included difficulty concentrating (72.4% of CD, 75.5% of NCGS); forgetfulness (60.3% of CD, 64.9% of NCGS); and grogginess (58.2% of CD, 69.2% of NCGS), among several other neurocognitive symptoms. Additionally, nearly 1/3rd (28% CD, 30% NCGS) of respondents indicated that they experienced these symptoms 1-4 hours post gluten exposure.

Respondents were asked to define their symptoms of "brain fog", and 32% elected to provide an open-ended response to this question.

This study sought to qualitatively analyze open-ended responses from the 2013 survey and identify themes common in reports of neurocognitive symptoms of gluten exposure in individuals with CD and NCGS.

These open-ended reflections illustrated a constellation of neurocognitive effects of gluten.

Methodology

All thematic coding and analyses were conducted in NVivo 11.

A coding scheme was developed by a team of experts for use in coding the neurocognitive symptoms reported by participants open-ended responses in the 2013 nationwide survey. The Health-Related Quality of Life Instrument (HRQOL) and a patient-derived conceptual model of the Impact of Celiac Disease in adults (Leffler et. al, 2017) were used as the foundation for the development of the neurocognitive fog coding scheme. Themes, such as physical and psychological symptoms, as well as impact on lifestyle and relationships, were incorporated into the neurocognitive fog coding structure.

Two trained raters reviewed and independently assigned codes to specific text references in the dataset. Any string of text could be coded, and coders were instructed to assign the most relevant code to a unique text string.

Leffler, D. A., Acaster, S., Gallop, K., Dennis, M., Kelly, C. P., & Adelman, D. C. (2017). A novel patient-derived conceptual model of the impact of celiac disease in adults: implications for patient-reported outcome and health-related quality-of-life instrument development. *Value in Health*, 20(4), 637-643.

Thematic Schema & Analysis

Coding categories (referred to as Nodes in Nvivo 11) derived from the development of the coding schema. Black nodes are parent nodes, which establish an overall thematic category. Red nodes are child nodes, which further refine the parent node (note: Child nodes 1.10 & 1.14 also contain child nodes). Gray nodes are child nodes that were not assigned to any text references by either coder. Parent nodes serve both as a codable node and an aggregate of itself and all child nodes.

1.0 Cognitive Impact	1.08 Spatial problem solving	1.13 Registration	2.0 Motor Impact	3.05 Bloating	3.13 Health related to celiac disease	4.01 Detached	6.0 Quality of Life Impact
1.01 Cognitive ability - IQ	1.09 Visual attention - vigilance	1.14 Attention	2.01 Psychomotor speed	3.06 Diarrhea	3.14 Health in general	4.02 Depressed	6.01 Daily functioning
1.02 Disinhibition	1.10 Memory	1.14a Attention and calculation	2.02 Basic motor skills	3.07 Sensation of not completely emptying bowels while going to the bathroom	3.15 Physical pain	4.03 Intoxicated	7.0 Time - Duration of Symptoms
1.03 Hyper-excitability	1.10a Visual learning - memory	1.14b Attention and working memory	3.0 Physical Impact	3.08 Hunger pains	3.16 Physical comfort	4.04 Anxious	8.0 Things that make Symptoms Better
1.04 Visual motor function	1.10b Verbal learning and memory	1.15 Language, impaired speech, expressive aphasia	3.01 Fatigue, grogginess, lethargy	3.09 Low energy	3.17 Sleep	4.05 Irritable, agitated	9.0 Things that make Symptoms Worse
1.05 Executive function	1.10c Recall	1.16 Repetition	3.02 Pain or discomfort in upper abdomen or the pit of the stomach	3.10 Headaches, migraines	3.18 Dizziness	5.0 Burden of Illness	
1.06 Psychomotor function	1.11 Orientation to time	1.17 Complex commands	3.03 Nausea	3.11 Food cravings	3.19 Balance	5.01 Medical burden	
1.07 Detection tasks	1.12 Orientation to place	1.18 Confusion	3.04 Rumbling in the stomach	3.12 Appetite	4.0 Psychological Impact	5.02 Social burden	

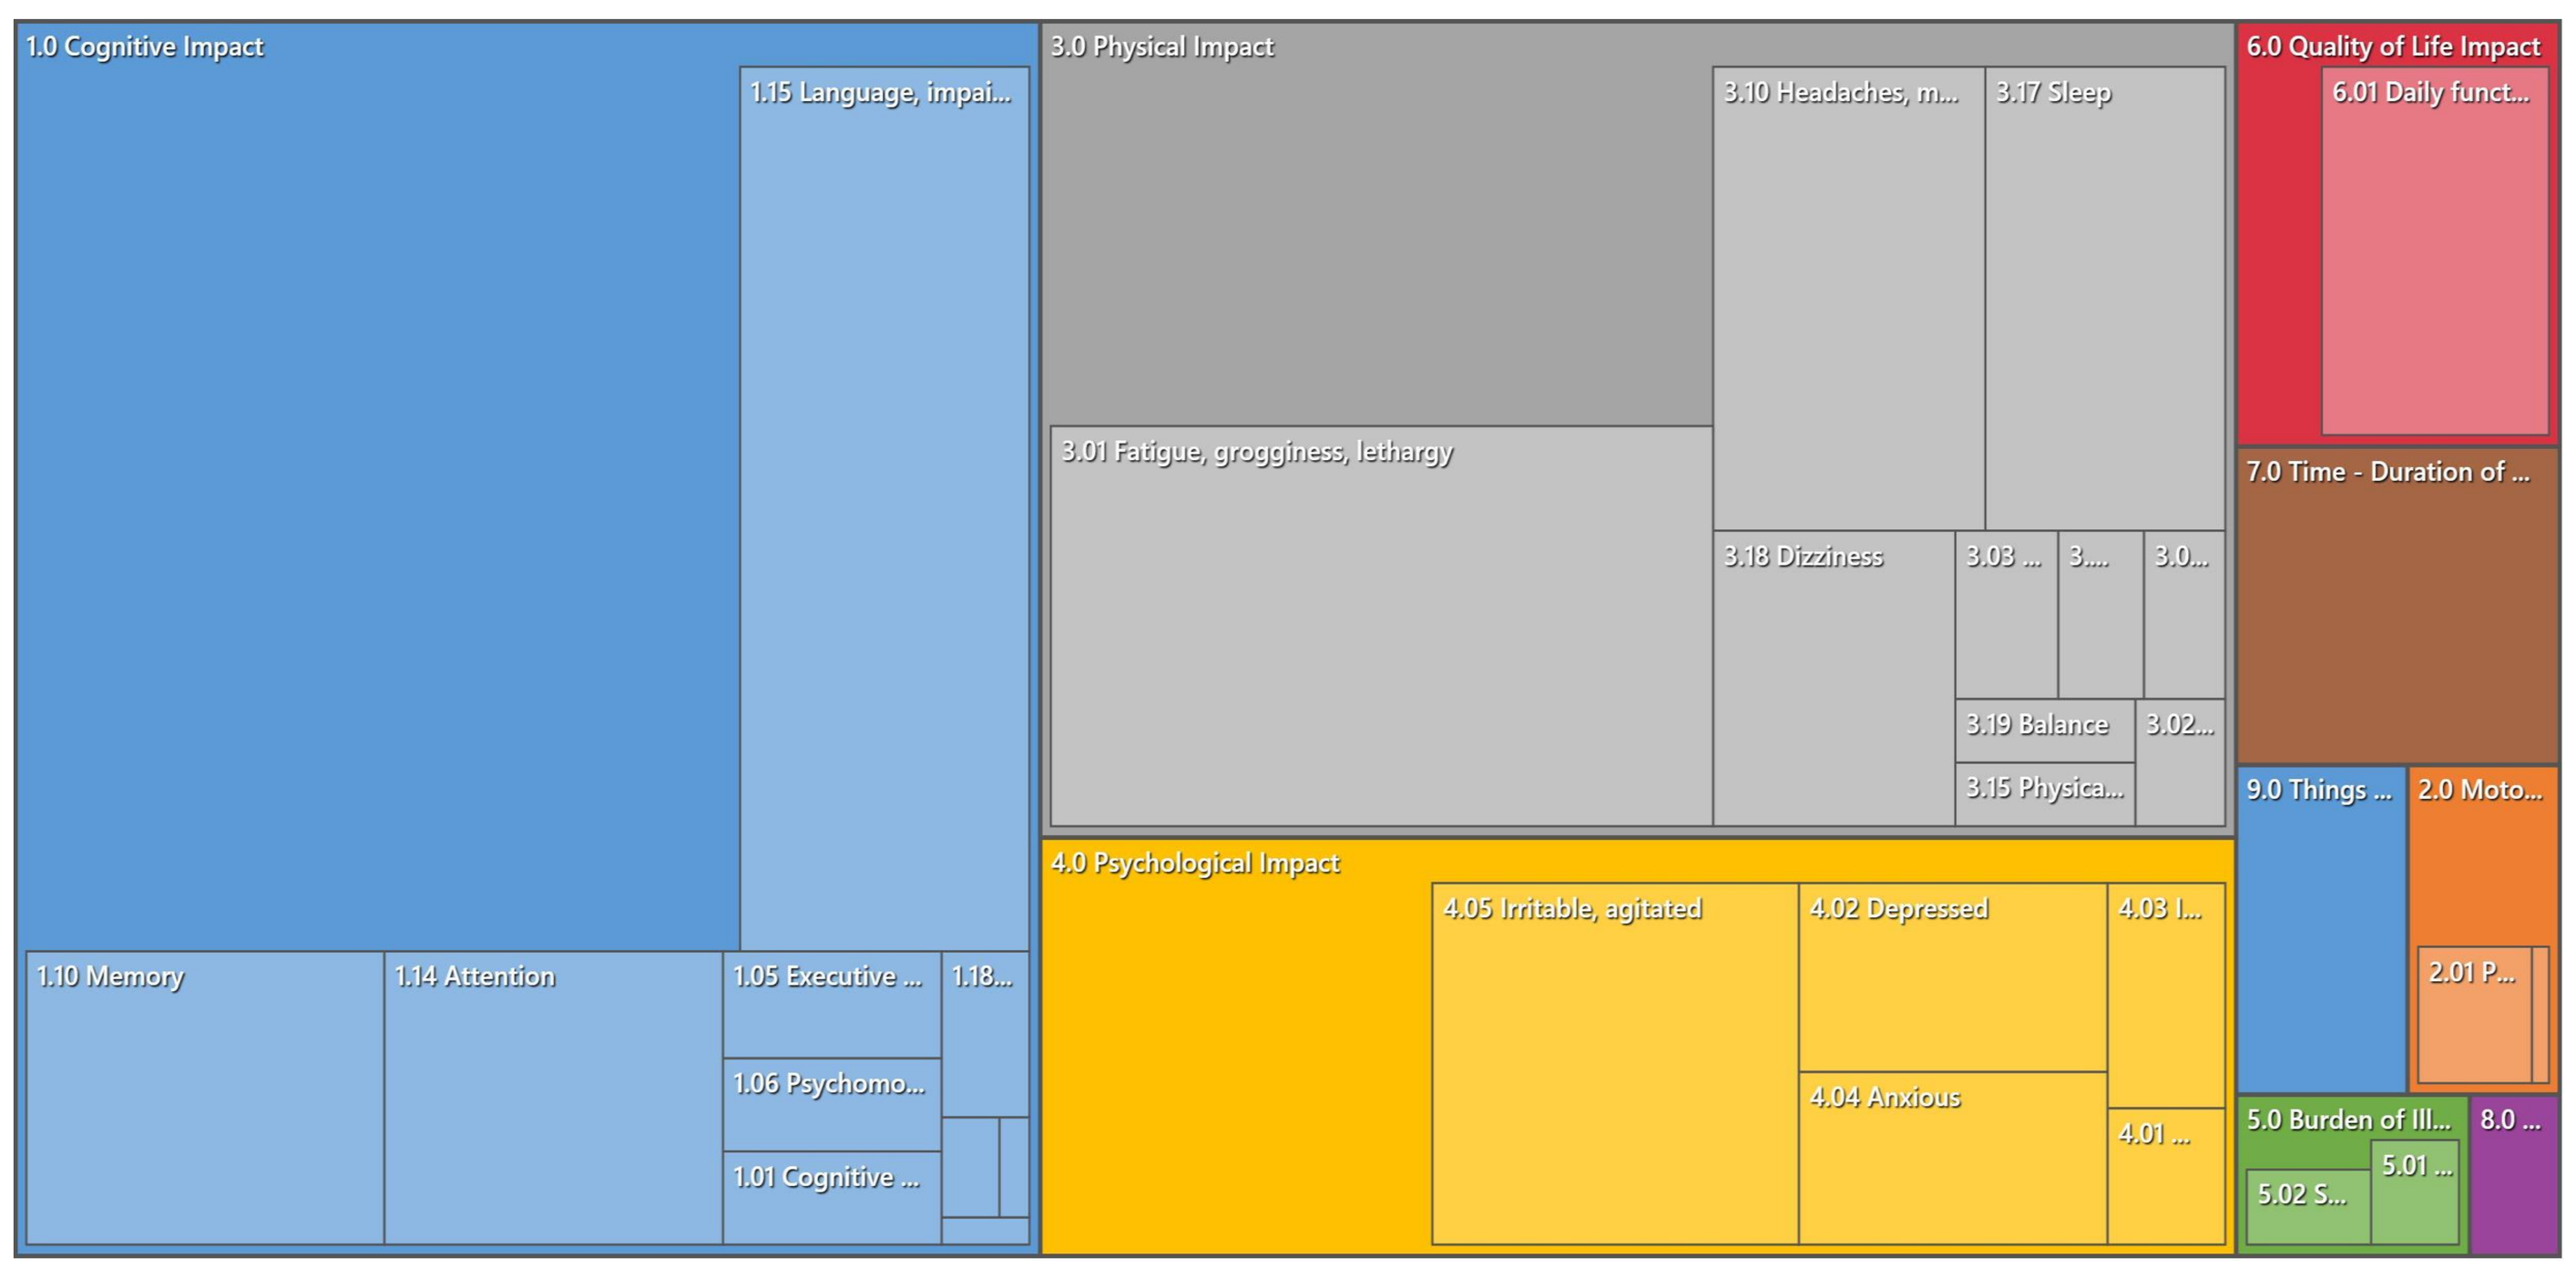
1.0 Cognitive Impact
1.15 Language, impai...
1.10 Memory
1.14 Attention
1.05 Executive ...
1.06 Psychomo...
1.01 Cognitive ...
3.0 Physical Impact
3.10 Headaches, m...
3.17 Sleep
3.01 Fatigue, grogginess, lethargy
3.18 Dizziness
3.19 Balance
3.15 Physica...
4.0 Psychological Impact
4.05 Irritable, agitated
4.02 Depressed
4.04 Anxious
6.0 Quality of Life Impact
6.01 Daily funct...
7.0 Time - Duration of ...
9.0 Things ...
2.0 Moto...
5.0 Burden of Ill...
8.0 ...

Above

Hierarchy of node coding relative to number of text references assigned to the node. Hierarchy of child nodes depicted within parent nodes.

Below

Table of top 10 parent two-set clusters. Nodes sets ranked by Pearson correlation coefficient.

Node A	Node B	Pearson correlation coefficient
9.0	7.0	0.45
6.0	1.0	0.42
9.0	8.0	0.35
4.0	3.0	0.32
3.0	1.0	0.30
9.0	6.0	0.22
9.0	1.0	0.21
8.0	7.0	0.21
4.0	1.0	0.21
9.0	3.0	0.20

Below

Cluster map of top 50 most common words clustered by co-occurrence (i.e. words that appear together or are near each other in the text). Words are clustered based on Pearson correlation coefficients.

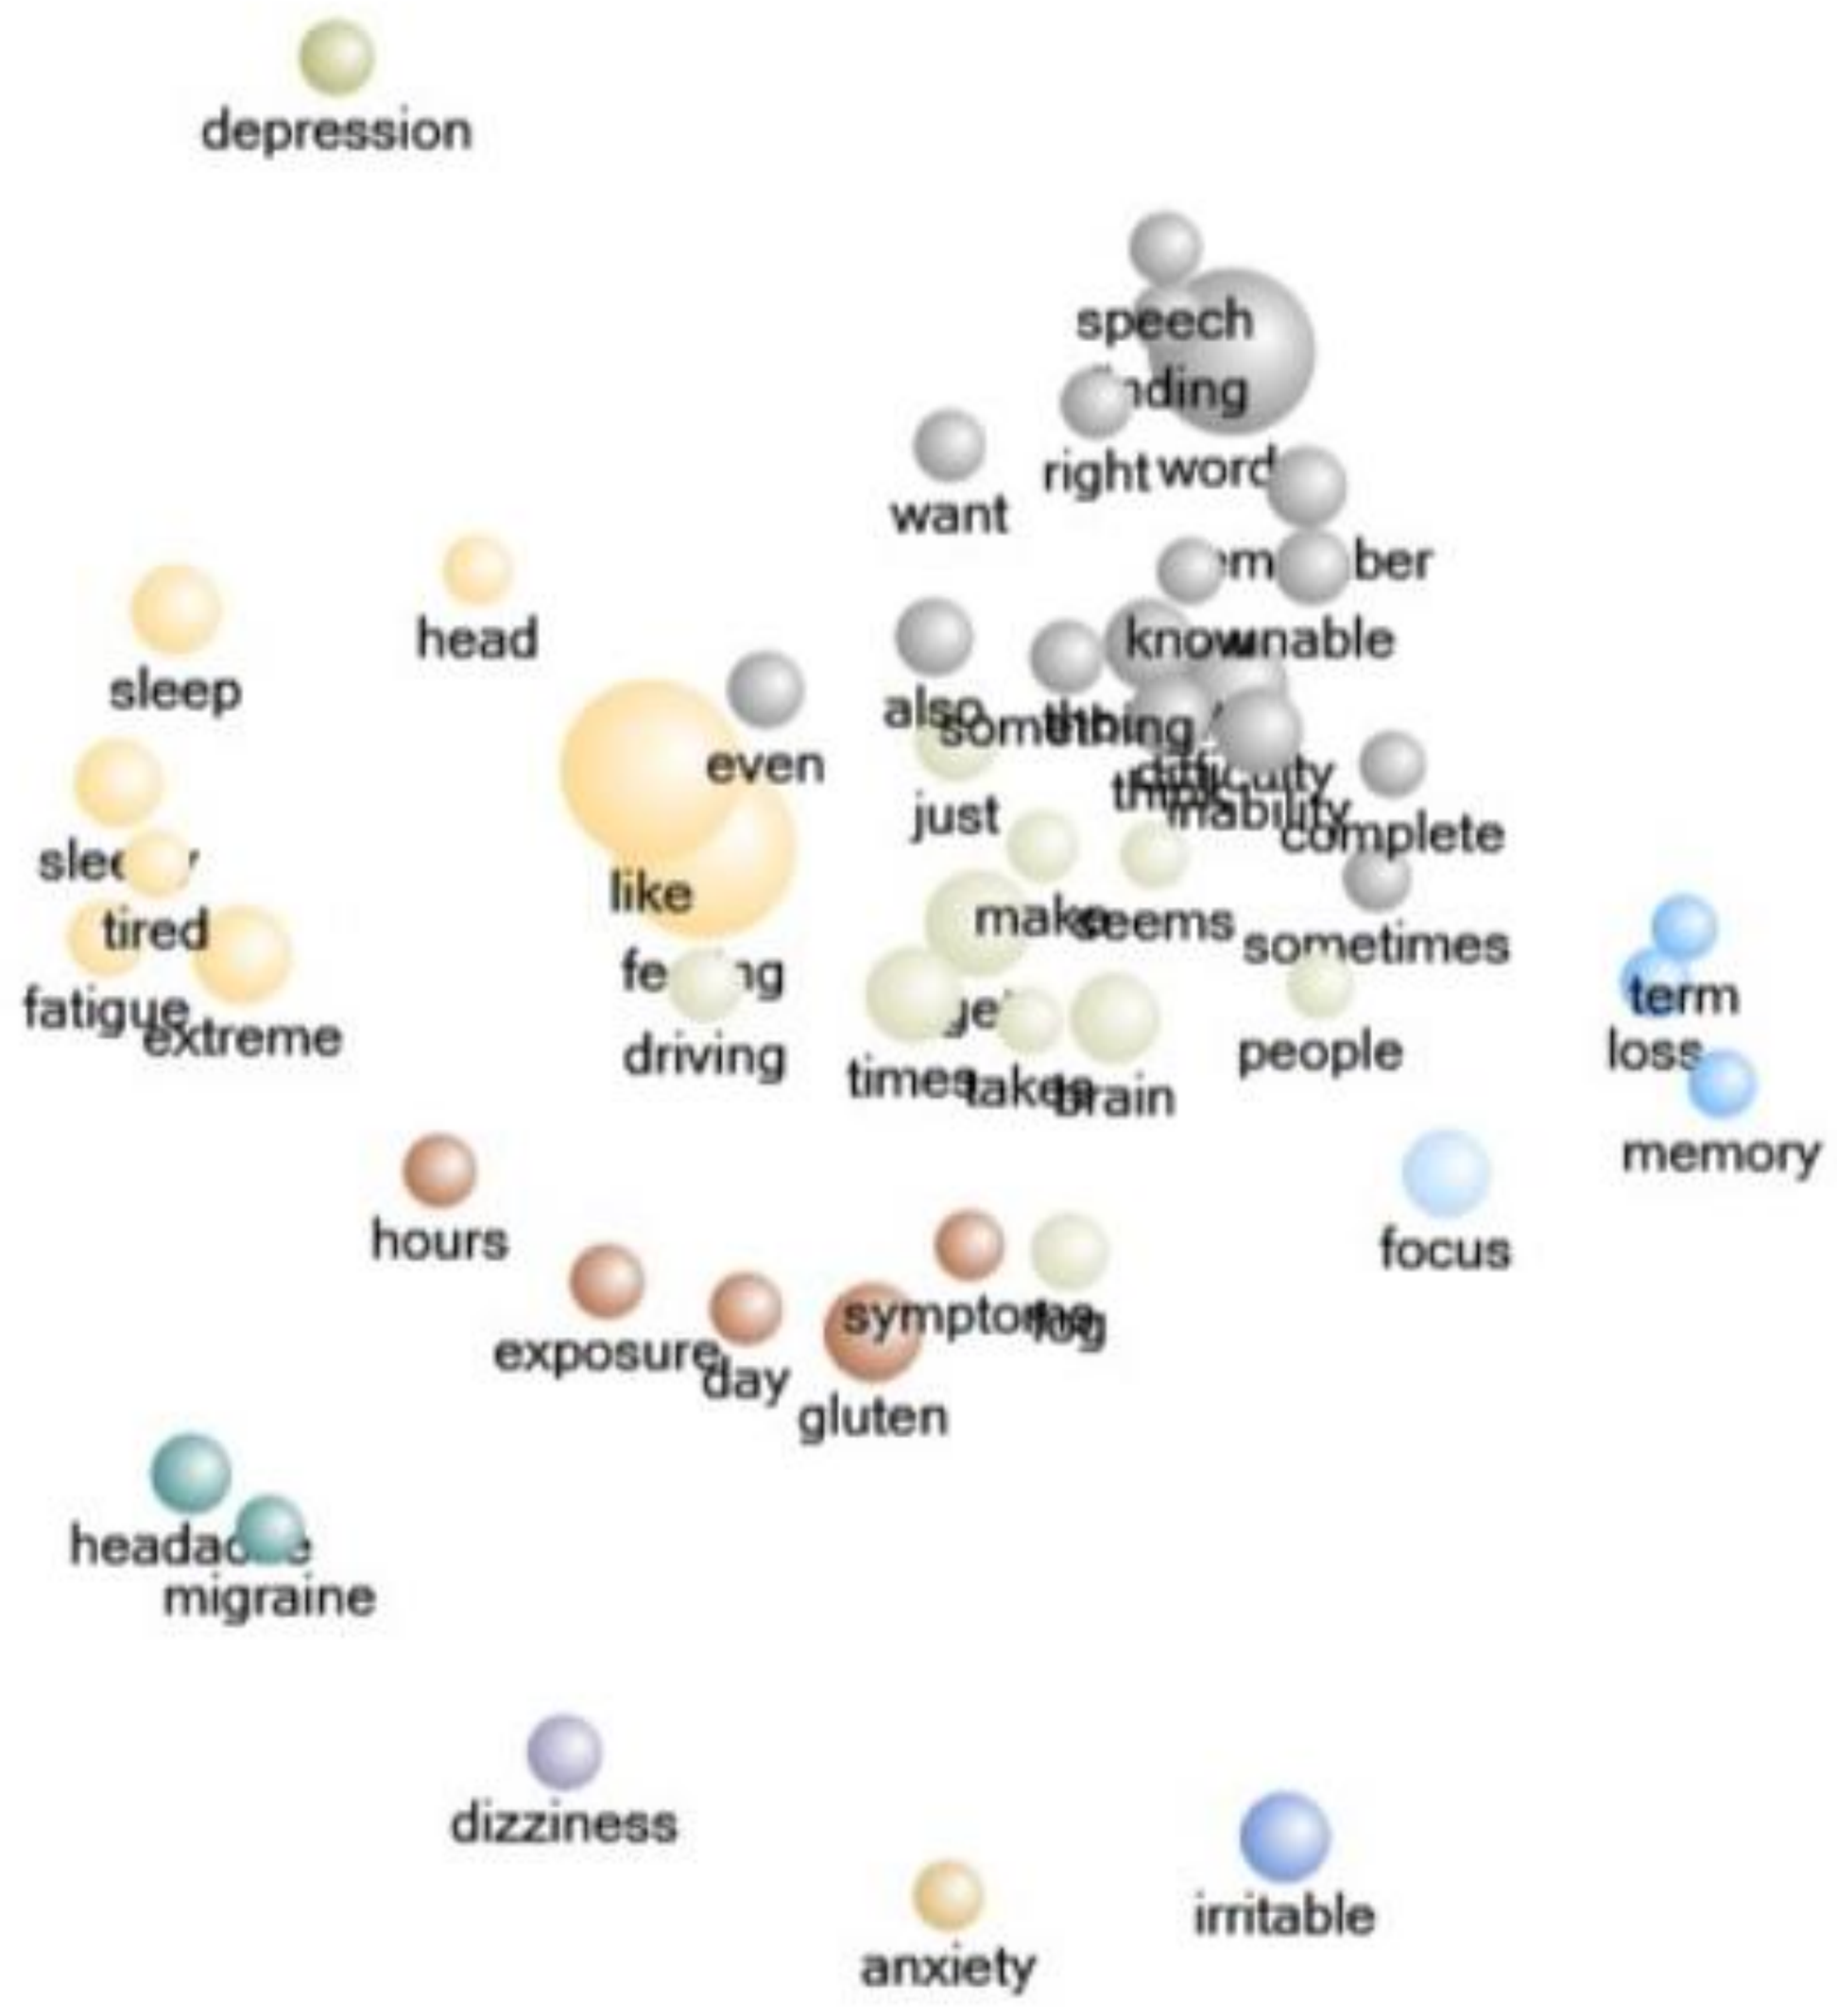
depression
speech
want
head
sleep
even
knownable
like
sometimes
people
term
loss
memory
tired
driving
hours
focus
exposure
gluten
migraine
dizziness
irritable
anxiety

Coding Comparison

Inter-rater reliability was analyzed in Nvivo 11. Text characters were the unit of analysis (total codable characters n = 28,541). Inter-rater reliability was evaluated with Cohen's Kappa, which accounts for random (by chance) agreement.

Note: only parent nodes are reported - parent nodes are an aggregate of the node itself and all child nodes.

Parent Node	Kappa	Agreement (%)	A and B (%)	Not A and Not B (%)	Disagreement (%)	A and Not B (%)	B and Not A (%)
All Nodes Average	0.63	93.04 %	6.94 %	86.10 %	6.96 %	1.92 %	5.04 %
1.0	0.66	83.22 %	36.84 %	46.38 %	16.78 %	8.74 %	8.04 %
2.0	0.43	98.66 %	0.52 %	98.14 %	1.34 %	0.00 %	1.34 %
3.0	0.65	88.99 %	14.13 %	74.87 %	11.01 %	3.41 %	7.60 %
4.0	0.40	92.17 %	3.06 %	89.11 %	7.83 %	0.93 %	6.90 %
5.0	0.02	94.70 %	0.12 %	94.58 %	5.30 %	1.78 %	3.52 %
6.0	0.13	89.31 %	0.96 %	88.35 %	10.69 %	0.61 %	10.08 %
7.0	0.68	95.54 %	5.24 %	90.30 %	4.46 %	1.28 %	3.17 %
8.0	0.85	99.80 %	0.58 %	99.21 %	0.17 %	0.00 %	0.17 %
9.0	0.27	94.93 %	1.02 %	93.91 %	5.07 %	0.52 %	4.54 %

Conclusion

Results show that the most referenced neurobehavioral and psychological symptoms were *cognitive* (node 1.0) , *physical* (3.0), *psychological* (4.0), and *QoL* (6.0) *impacts*. These nodes may assist healthcare providers and patients to describe neurocognitive fog symptoms with increased ease allowing for targeted treatment of symptoms and their link with adherence/gluten exposure. *Cognitive impact* (1.0) and *QoL impact* (6.0) node sets were found to be moderately correlated. This result provides potential insight into the impact of neurobehavioral symptoms on QoL and aligns with common frameworks for evaluating neurocognitive symptoms (e.g. declines in both cognitive functioning and ADLs). Preliminary results of the coding comparison show moderate agreement across all nodes, with the highest agreement with the node *things that make symptoms* better (8.0). Future directions are to reconcile textual and thematic coding disagreements to mastery, and further evaluate what may actually be an underestimated IRR.

BEYOND CELIAC

awareness • advocacy • action

Takeda

LVX VERITAS VIRTVS